Week 24 Outcomes of F/TAF Plus Cobicistat-Boosted Protease Inhibitors in Children ≥ 2 Years and ≥ 14 kg

1049

GS-US-216-0128

Hilda Angela Mujuru[1], **Renate Strehlau**[2], **Pope Kosalaraksa**[3], **Jaime Gerardo Deville**[4], **Meiling Pan**[5], **Vinicius Adriano Vieira**[5], **Kathryn Kersey**[5], **Natella Rakhmanina**[6]

[1]University of Zimbabwe, Harare, Zimbabwe; [2]University of the Witwatersrand, Johannesburg, South Africa; [3]Khon Kaen University, Khon Kaen, Thailand; [4]Mattel Children's Hospital, University of California, Los Angeles, CA, USA; [5]Gilead Sciences, Inc., Foster City, CA, USA; [6]Children's National Hospital, The George Washington University, Washington, DC, USA

Conclusions

- In this Week 24 analysis in children aged 2 to < 12 years and weighing 14 to < 40 kg, F/TAF in combination with ATV/c or DRV/c demonstrated favorable efficacy and safety
- Most participants maintained or achieved virologic suppression through 24 weeks of treatment
- F/TAF in combination with ATV/c or DRV/c was well tolerated
 - There were no renal, bone, or metabolic safety concerns
- These data support further evaluation of F/TAF in combination with ATV/c or DRV/c in children with HIV
 - Additional analyses will be conducted after Week 48 of treatment

Plain Language Summary

- F/TAF is a single tablet containing two different medicines used to treat human immunodeficiency virus (HIV): emtricitabine (F) and tenofovir alafenamide (TAF)
 - It is normally taken with a third medicine
- F/TAF has been approved to be used in combination with medicines called boosted protease inhibitors in children and teenagers who weigh at least 35 kg (77 lb)
 - We are now carrying out studies with F/TAF in younger children
- In this study, children aged 2 years and over who weigh at least 14 kg (31 lb) are taking F/TAF plus either cobicistat-boosted atazanavir (ATV/c) or cobicistat-boosted darunavir (DRV/c)
- This poster reports results after 24 weeks, showing how well the medicines are working, if there are any side effects, and how easy the tablets are to take
- After 24 weeks, F/TAF with ATV/c or DRV/c worked well at controlling the amount of HIV in the blood
- Side effects were rare, and the tablets were easy to take
- The study will now continue to collect more results after 48 weeks of treatment

Introduction

- Nucleoside/nucleotide reverse transcriptase inhibitors (NRTIs) in combination with a boosted protease inhibitor (PI) are a recommended treatment for children with HIV-1 who have resistance or intolerability to integrase strand transfer inhibitors[1]
- Emtricitabine/tenofovir alafenamide (F/TAF) is a dual NRTI approved in the US for use in combination with boosted PIs for adults, and for children and adolescents weighing ≥ 35 kg, and with other antiretrovirals (ARVs) for children weighing ≥ 14 kg[1,2]; it is approved in the European Union in combination with other ARVs, including boosted PIs, for adults and adolescents aged ≥ 12 years and weighing ≥ 35 kg[3]
 - TAF has improved renal and bone safety compared with tenofovir disoproxil fumarate (TDF)[4]
 - Cobicistat is a pharmacokinetic enhancer with no antiretroviral activity that can be easily coformulated with other ARVs[5]
- Safety and efficacy data for cobicistat-boosted PIs, including boosted PIs in combination with F/TAF in the pediatric population, are limited
- GS-US-216-0128 (NCT02016924) is an ongoing Phase 2/3, multicenter, open-label, multicohort trial evaluating F/TAF and boosted PIs in children and adolescents with HIV-1

Objective

- To evaluate the efficacy and safety of F/TAF in combination with cobicistat-boosted atazanavir (ATV/c) or darunavir (DRV/c) in children with HIV-1 aged 2 to < 12 years and weighing 14 to < 40 kg from Cohorts 2 and 3 of Study GS-US-216-0128 (NCT02016924) through Week 24

Methods

- This analysis focused on participants who were aged 6 to < 12 years weighing 25 to < 40 kg (Cohort 2) and aged ≥ 2 years weighing 14 to < 25 kg (Cohort 3)

Study Design

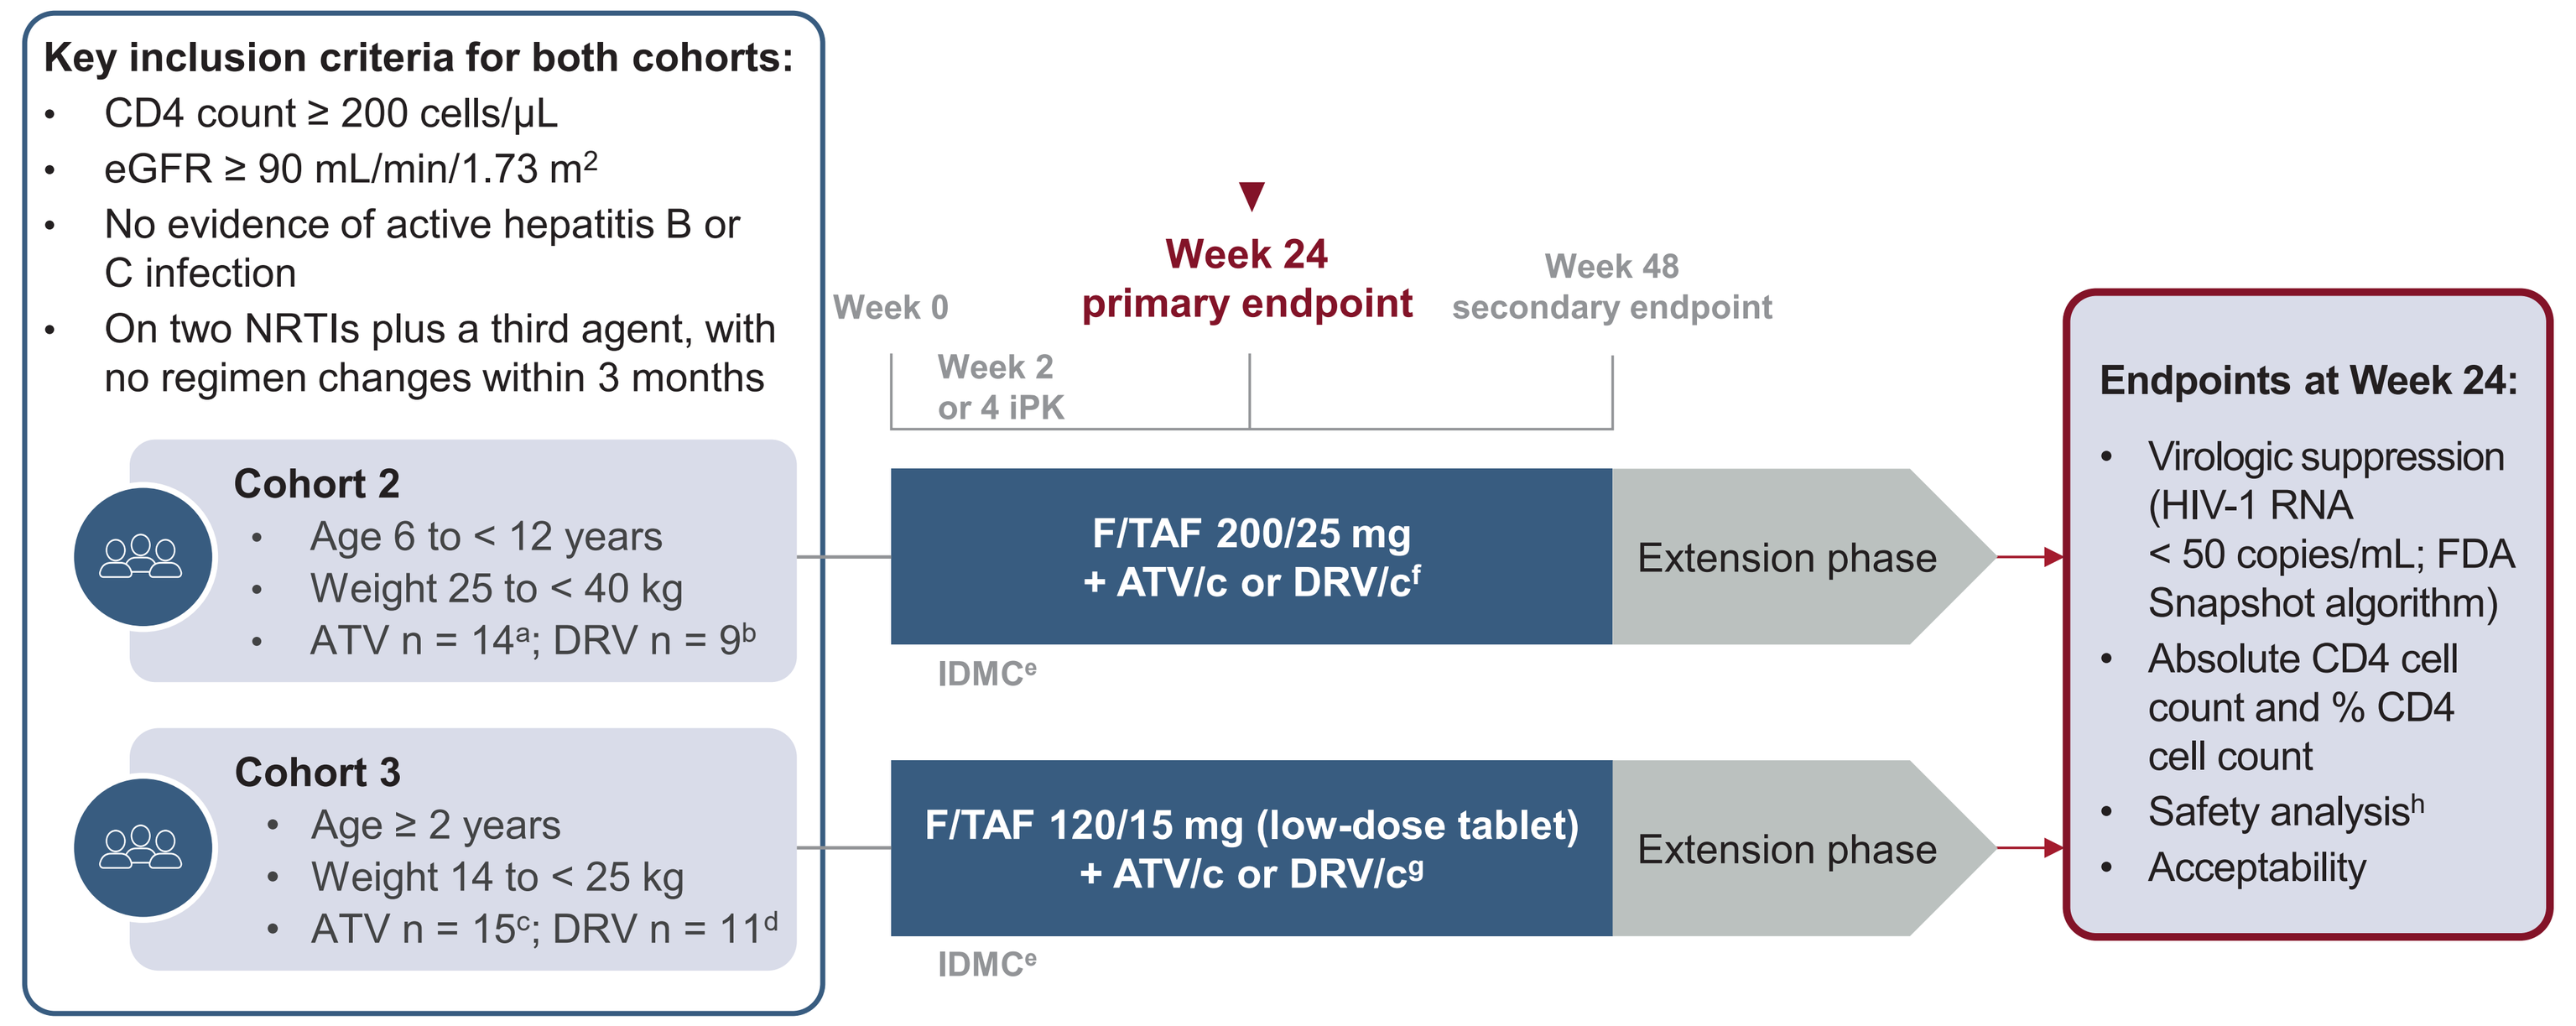

Key inclusion criteria for both cohorts:
CD4 count ≥ 200 cells/μL
eGFR ≥ 90 mL/min/1.73 m²
No evidence of active hepatitis B or C infection
On two NRTIs plus a third agent, with no regimen changes within 3 months
Cohort 2
Age 6 to < 12 years
Weight 25 to < 40 kg
ATV n = 14a; DRV n = 9b
Cohort 3
Age ≥ 2 years
Weight 14 to < 25 kg
ATV n = 15c; DRV n = 11d
Week 0
Week 2 or 4 iPK
Week 24 primary endpoint
Week 48 secondary endpoint
F/TAF 200/25 mg + ATV/c or DRV/cf
F/TAF 120/15 mg (low-dose tablet) + ATV/c or DRV/cg
Extension phase
IDMCe
Endpoints at Week 24:
Virologic suppression (HIV-1 RNA < 50 copies/mL; FDA Snapshot algorithm)
Absolute CD4 cell count and % CD4 cell count
Safety analysish
Acceptability

Enrollment: [a]South Africa n = 8, Zimbabwe n = 6. [b]South Africa n = 3, USA n = 1, Zimbabwe n = 5. [c]South Africa n = 10, Zimbabwe n = 5. [d]South Africa n = 7, Zimbabwe n = 4. [e]Data review by the IDMC occurred after the last participant was enrolled in each cohort and ≥ 50% of participants had completed Week 12. [f]All participants weighing ≥ 35 kg receive DRV; cobicistat dose is 150 mg; ATV and DRV are dosed by weight. [g]Participants must be aged ≥ 3 years and weigh ≥ 15 kg to receive DRV; cobicistat is a 90-mg low-dose tablet; ATV and DRV are dosed by weight. [h]Cumulative through data-cut (when the last participant enrolled in Cohorts 2 and 3 had completed Week 24).
ATV, atazanavir; c, cobicistat; CD4, cluster of differentiation 4; DRV, darunavir; eGFR, estimated glomerular filtration rate by Schwartz formula; F, emtricitabine; IDMC, Independent Data Monitoring Committee; iPK, intensive pharmacokinetics; NRTI, nucleoside/nucleotide reverse transcriptase inhibitor; TAF, tenofovir alafenamide.

Results

Baseline Demographics and Disease Characteristics

	Cohort 2 (6 to < 12 years, 25 to < 40 kg) n = 23	Cohort 3 (≥ 2 years, 14 to < 25 kg) n = 26
Age, years, median (range)	10 (8-12)	6 (3-10)
Female sex at birth, n (%)	14 (61)	14 (54)
Race, n (%)		
Black	21 (91)	24 (92)
Other	2[a] (9)	2[b] (8)
Hispanic or Latine ethnicity,[c] n/N (%)	1/22 (5)	1/25 (4)
HIV-1 RNA < 50 c/mL, n (%)	22 (96)	24 (92)
CD4 count, cells/μL, median (Q1, Q3)	876 (671, 1063)	940 (705, 1259)
CD4%, median (Q1, Q3)	36.2 (31.1, 41.0)	34.8 (29.5, 40.8)
Vertical transmission, n (%)	22[d] (96)	26 (100)
Asymptomatic disease status, n (%)	22 (96)	26 (100)

[a]One participant was of mixed race (Black/White) and one participant did not want to report their race. [b]Both participants were of mixed race (Black/White). [c]Data were unavailable for one participant in each cohort. [d]Mode of transmission was unknown for one participant.
c, copies; CD4, cluster of differentiation 4; Q, quartile.

Efficacy Outcomes at Week 24

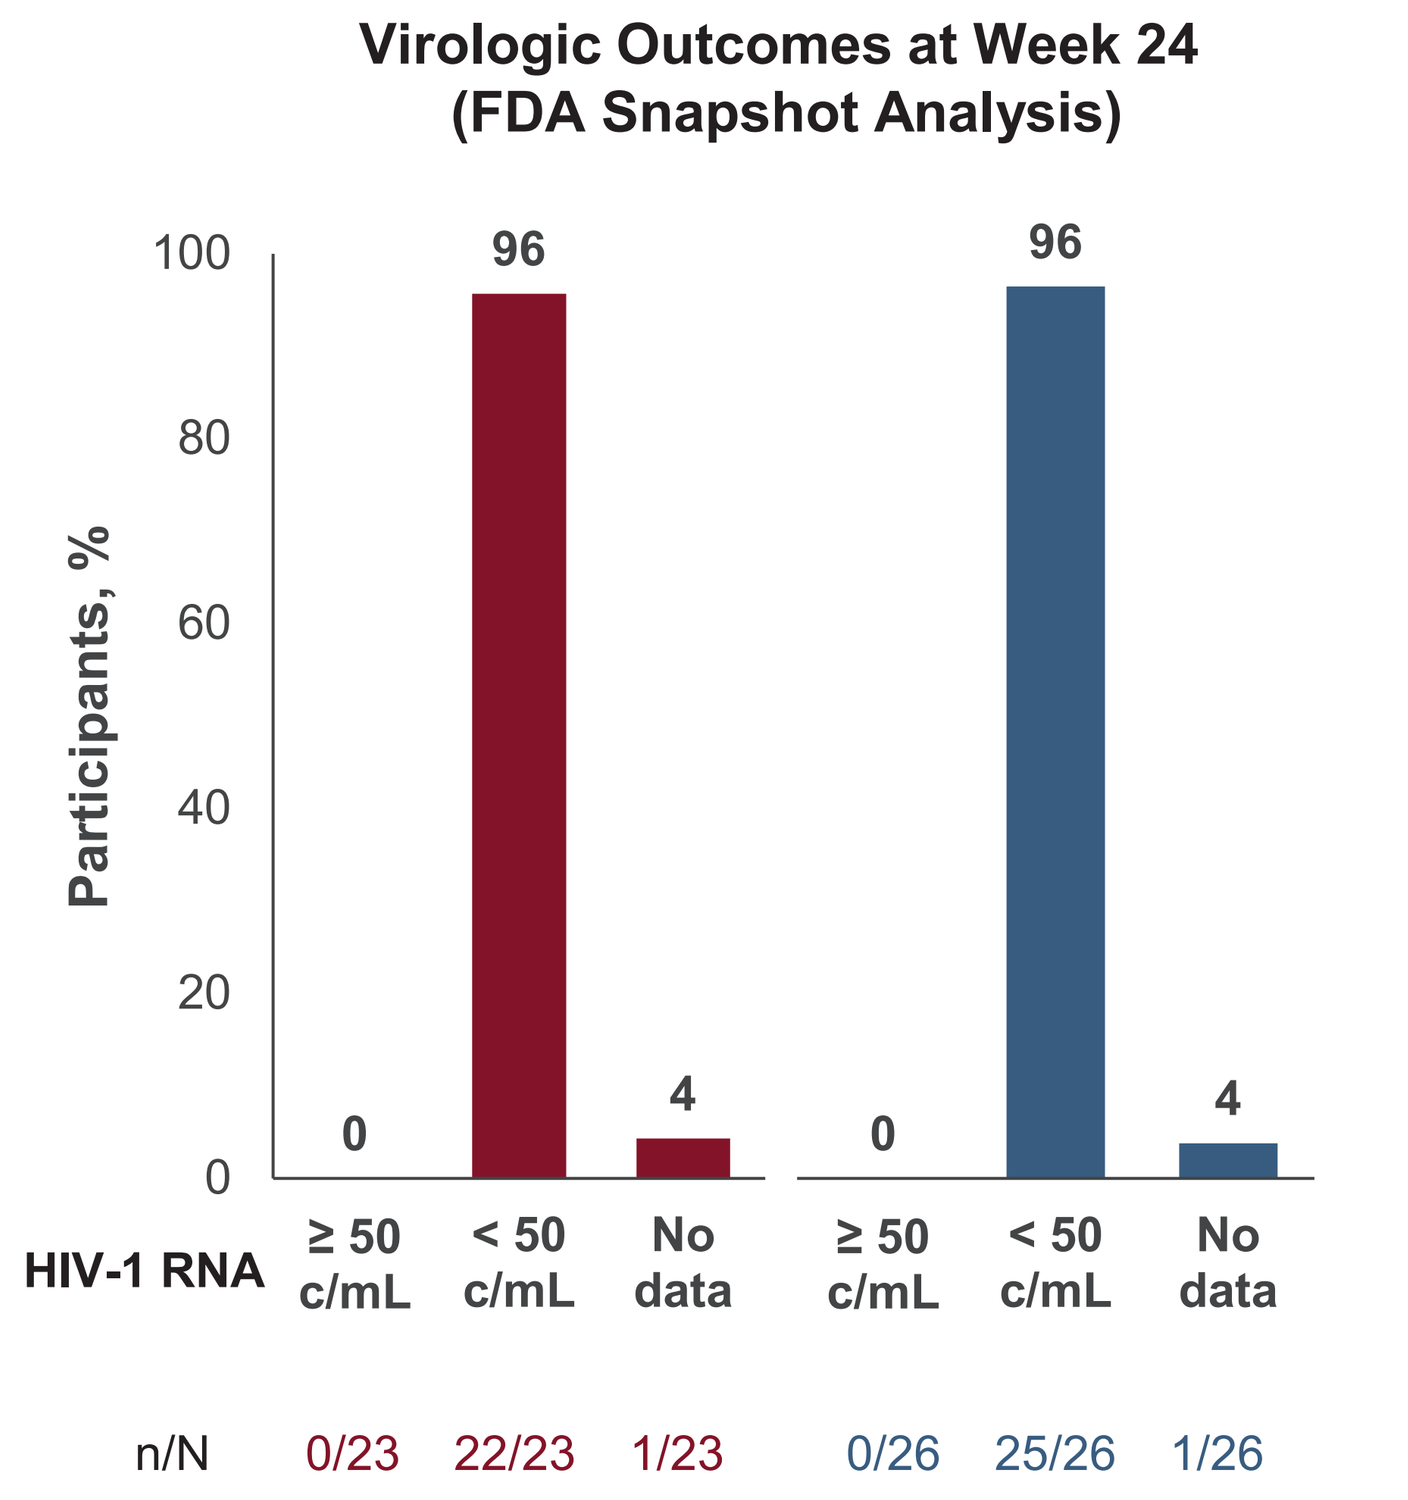

Virologic Outcomes at Week 24 (FDA Snapshot Analysis)
Participants, %
100
80
60
40
20
0
96
0
4
96
0
4
HIV-1 RNA
≥ 50 c/mL
< 50 c/mL
No data
≥ 50 c/mL
< 50 c/mL
No data
n/N
0/23
22/23
1/23
0/26
25/26
1/26

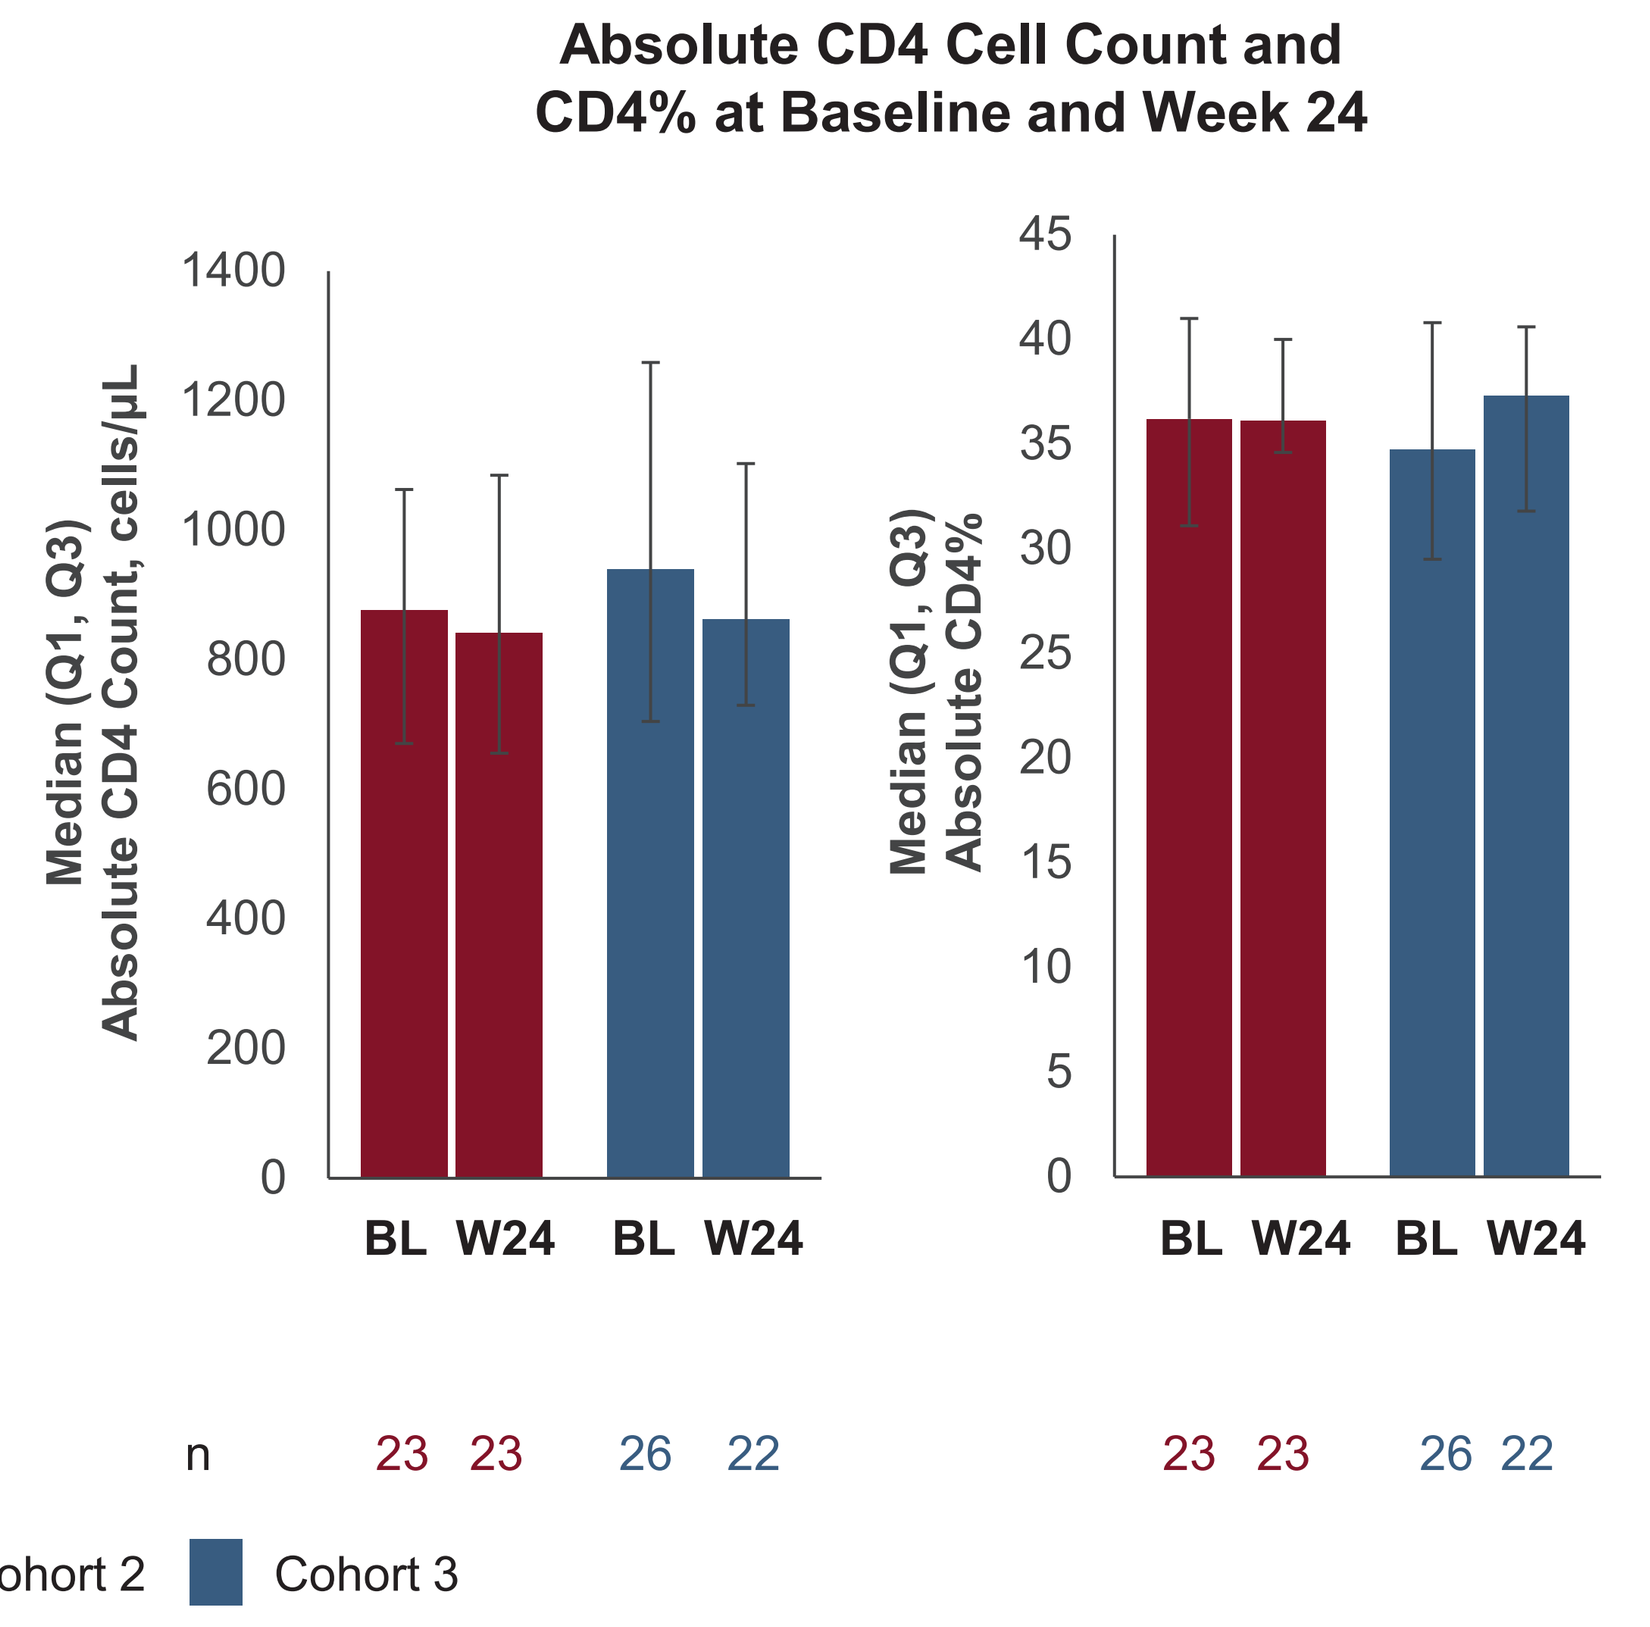

Absolute CD4 Cell Count and CD4% at Baseline and Week 24
Median (Q1, Q3) Absolute CD4 Count, cells/μL
1400
1200
1000
800
600
400
200
0
Median (Q1, Q3) Absolute CD4%
45
40
35
30
25
20
15
10
5
0
BL W24 BL W24
BL W24 BL W24
n
23 23 26 22
23 23 26 22
Cohort 2
Cohort 3

Cohort 2: 6 to < 12 years, 25 to < 40 kg; Cohort 3: ≥ 2 years, 14 to < 25 kg.
BL, baseline; c, copies; CD4, cluster of differentiation 4; Q, quartile; W, Week.

- Most participants maintained or achieved virologic suppression at Week 24
- Absolute CD4 cell count and percentage remained stable

Safety Outcomes

n (%)	Cohort 2 (6 to < 12 years, 25 to < 40 kg) n = 23		Cohort 3 (≥ 2 years, 14 to < 25 kg) n = 26	
	ATV/c + F/TAF n = 14	DRV/c + F/TAF n = 9	ATV/c + F/TAF n = 15	DRV/c + F/TAF n = 11
Any AE[a]	11 (79)	8 (89)	12 (80)	11 (100)
DRAEs[b]	4 (29)	3 (33)	5 (33)	3 (27)
Grade 3/4 DRAEs	0	0	2 (13)	0
Increased bilirubin/hyperbilirubinemia[c]	0	0	2 (13)	0
Serious DRAEs	0	0	1 (7)	0
Hyperbilirubinemia[c]	0	0	1 (7)	0
AEs leading to study drug discontinuation	4 (29)	0	1 (7)	0
Deaths	0	0	0	0

Safety outcomes are cumulative over the duration of the study.
[a]AEs experienced by > 10% of participants overall were: URTI n = 15 (31%), vomiting n = 10 (20%), and hyperbilirubinemia n = 5 (10%). [b]DRAEs experienced by participants in Cohort 2 receiving ATV were: hyperbilirubinemia n = 2 (14%), vomiting n = 1 (7%), increased blood bilirubin n = 1 (7%), and ocular icterus n = 1 (7%); DRAEs experienced by participants in Cohort 2 receiving DRV were: vomiting n = 2 (22%) and headache n = 1 (11%); DRAEs experienced by participants in Cohort 3 receiving ATV were: hyperbilirubinemia n = 3 (20%), URTI n = 1 (7%), abnormal blood bilirubin n = 1 (7%), and seasonal allergy n = 1 (7%); DRAEs experienced by participants in Cohort 3 receiving DRV were: vomiting n = 2 (18%), URTI n = 1 (9%), abdominal pain n = 1 (9%), and fungal skin infection n = 1 (9%). [c]All considered related to ATV by the investigators.
AE, adverse event; ATV, atazanavir; c, cobicistat; DRAE, drug-related adverse event; DRV, darunavir; F/TAF, emtricitabine/tenofovir alafenamide; URTI, upper respiratory tract infection.

- Median (quartile [Q]1, Q3) duration of study drug treatment was 96.1 (35.0, 141.0) and 120.1 (64.1, 160.0) weeks for Cohorts 2 and 3, respectively[d]
- Grade 3/4 laboratory abnormalities in > 1 participant were increased bilirubin (Cohort 2: n = 7 [30%]; Cohort 3: n = 6 [23%]), increased amylase (Cohort 2: n = 5 [22%]; Cohort 3: n = 4 [15%]), and decreased neutrophils (Cohort 2: n = 3 [13%]; Cohort 3: n = 3 [12%])
- All reports of increased bilirubin were considered related to ATV by the investigators
- Four participants, all receiving ATV, switched to DRV after the Week 24 visit because of hyperbilirubinemia[e]

[d]The large variation in time on treatment was related to slow enrollment, which is common in pediatric studies; all available safety data are included in this analysis.
[e]Participants were permitted to switch from ATV to DRV if they experienced drug-related clinically significant hyperbilirubinemia.

Height, Weight, Bone, and Renal Parameters at Baseline and Week 24

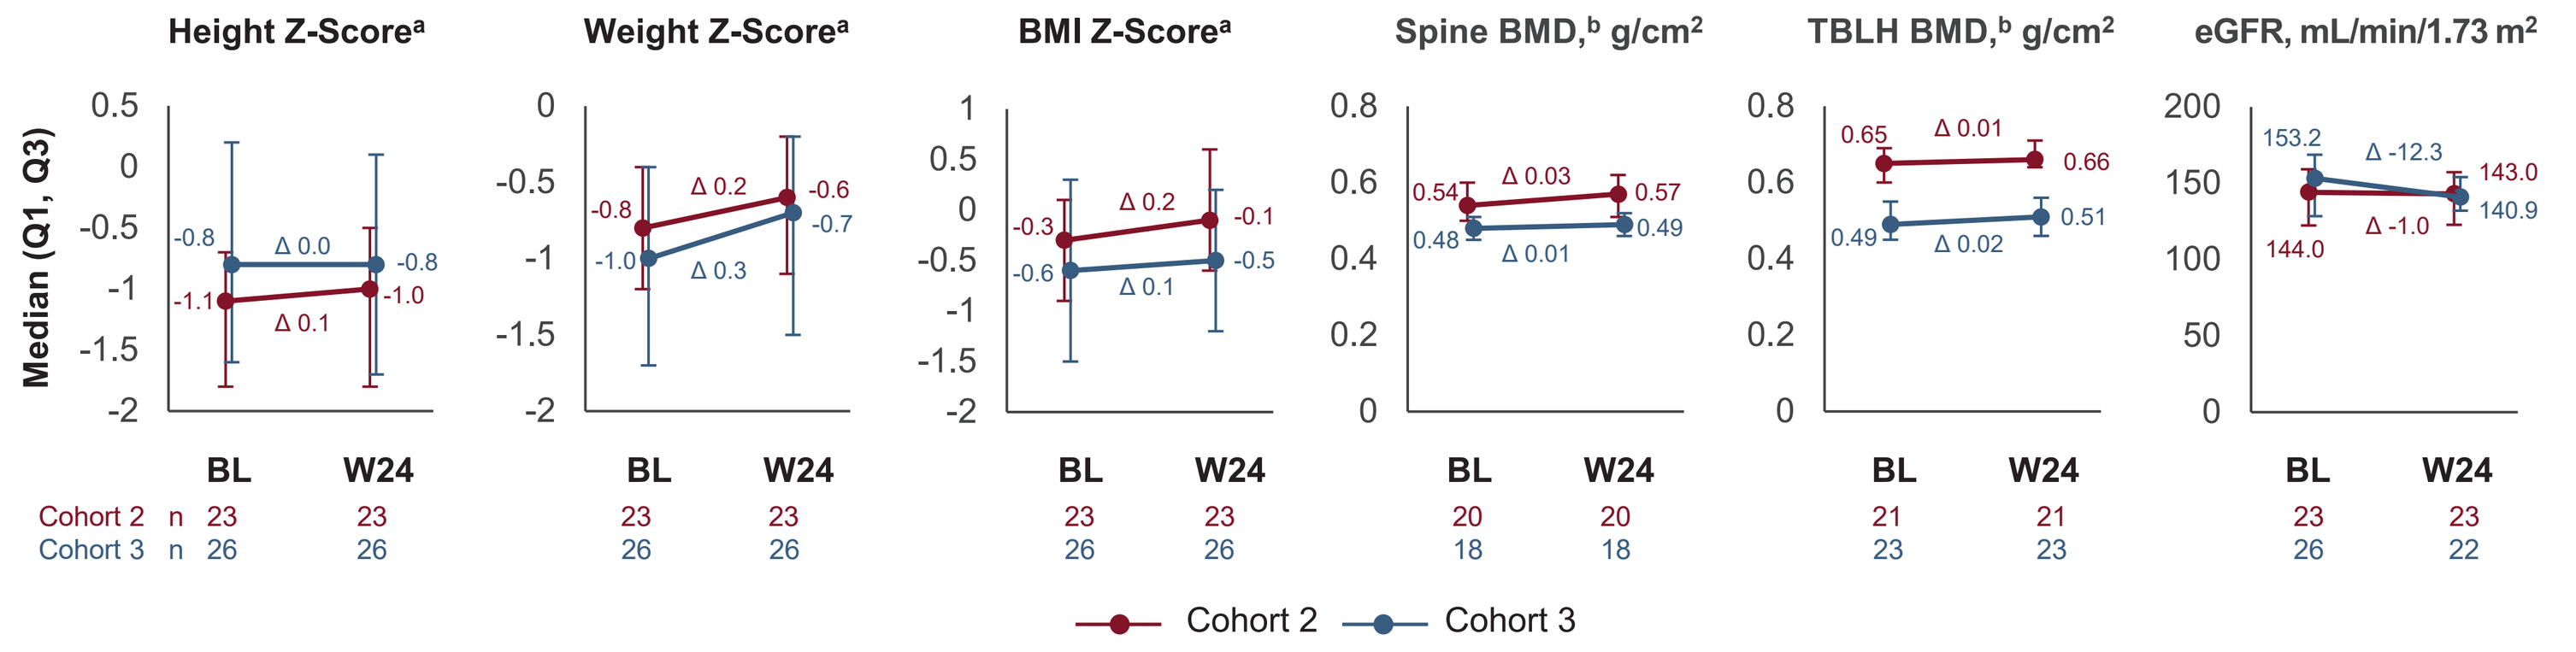

Height Z-Scorea
Weight Z-Scorea
BMI Z-Scorea
Spine BMD,b g/cm²
TBLH BMD,b g/cm²
eGFR, mL/min/1.73 m²
Median (Q1, Q3)
-0.8 Δ 0.0 -0.8
-1.1 Δ 0.1 -1.0
-0.8 Δ 0.2 -0.6
-1.0 Δ 0.3 -0.7
-0.3 Δ 0.2 -0.1
-0.6 Δ 0.1 -0.5
0.54 Δ 0.03 0.57
0.48 Δ 0.01 0.49
0.65 Δ 0.01 0.66
0.49 Δ 0.02 0.51
153.2 Δ -12.3 140.9
144.0 Δ -1.0 143.0
BL W24
Cohort 2 n 23 23 | 23 23 | 23 23 | 20 20 | 21 21 | 23 23
Cohort 3 n 26 26 | 26 26 | 26 26 | 18 18 | 23 23 | 26 22
Cohort 2
Cohort 3

Cohort 2: 6 to < 12 years, 25 to < 40 kg; Cohort 3: ≥ 2 years, 14 to < 25 kg. Baseline values were the last available values collected on/before first dose of study drug. [a]Z-scores generated using 2000 US Centers for Disease Control and Prevention Growth Charts. [b]BMD assessed using dual-energy X-ray absorptiometry of spine and total body less head.
BL, baseline; BMD, bone mineral density; BMI, body mass index; eGFR, estimated glomerular filtration rate by Schwartz formula; Q, quartile; TBLH, total body less head; W, Week.

- There were no clinically significant changes in height, weight, bone, and renal parameters from baseline to Week 24

Acceptability

- In Cohort 2 at Week 24, 100% (22/22) of participants with available data reported the study drugs to have an acceptable size and shape when swallowed whole
- In Cohort 3 at Week 24, 100% (25/25) of participants with available data reported that the size of the low-dose F/TAF tablet was "okay" and 100% (26/26) reported that it was "easy"/"super easy" to swallow; 96% (25/26) reported that the tablet had a "good"/"super good" shape to swallow

References: 1. Department of Health and Human Services. https://clinicalinfo.hiv.gov/sites/default/files/guidelines/documents/pediatric-arv/guidelines-pediatric-arv.pdf (accessed Nov. 20, 2024). 2. Descovy USPI, Gilead Sciences, Inc., January 2022. 3. Descovy SmPC, Gilead Sciences, Inc., February 2023. 4. DeJesus E, et al. *AIDS Res Hum Retroviruses.* 2018;34:337-42. 5. Clinicalinfo.HIV.gov. https://clinicalinfo.hiv.gov/en/drugs/cobicistat/patient (accessed Nov. 20, 2024).

Acknowledgments: We thank all study participants, study investigators, and staff. We also thank Brenda Okware (Gilead Sciences, Inc.) for reviewing the presentation. This study was sponsored by Gilead Sciences, Inc. Medical writing support was provided by Lindsay Fawcett, BSc (Aspire Scientific Ltd, UK) and was funded by Gilead Sciences, Inc.

Disclosures: **RS** reports research funding paid to their institution by Gilead Sciences, Inc., consulting fees for advisory boards for GSK and ViiV Healthcare, and support for attending meetings and/or travel from Gilead Sciences, Inc. and Enanta Pharmaceuticals. **PK** and **JGD** report research funding paid to their institution by Gilead Sciences, Inc. **MP** is a contracted study statistician from The Lotus Group, which receives payment from Gilead Sciences, Inc. **VAV** and **KK** are employees of, and own stocks/shares in, Gilead Sciences, Inc. **NR** reports research funding paid to their institution from Gilead Sciences, Inc., GSK, Merck, and Penta; personal payment for expert testimony on an advisory board from ViiV Healthcare; and travel support from Gilead Sciences, Inc. **HAM** has no conflicts of interest to report.

Correspondence: Hilda Angela Mujuru, hmujuru@mweb.co.zw.

Conference on Retroviruses and Opportunistic Infections (CROI); March 9–12, 2025; San Francisco, CA, USA